757

Vancomycin-resistant enterococcal (VRE) titers diminish among patients with recurrent Clostridium difficile infection after administration of SER-109, a novel microbiome agent

Mary-Jane Lombardo, PhD[1], Marin Vulic, PhD[1], Toshi Ohsumi, PhD[1], David Cook, PhD[1], Matthew Henn, PhD[1], Colleen Kraft, MD[2], Sahil Khanna, MBBS, MS[3], Darrell Pardi, MD, MS[3] and Elizabeth Hohmann, MD, FIDSA[4]

(1)Seres Therapeutics, Inc, Cambridge, MA, (2)Emory University School of Medicine, Atlanta, GA, (3)Division of Gastroenterology and Hepatology, Mayo Clinic, Rochester, MN, (4)Infectious Diseases, Massachusetts General Hospital, Boston, MA

Seres Therapeutics, Inc.
215 First Street, Suite 100
Cambridge, MA 02142
617-945-9626

Abstract

Background: VRE infections are a worldwide problem due to increasing prevalence, lack of effective therapies and modest impact of infection control policies. A leading risk factor for VRE is prior antibiotic use, which disrupts the healthy microbiome. In a study of patients with recurrent *Clostridium difficile* infection (RCDI), we assessed the prevalence of VRE before and after administration of SER-109, a novel microbiome-based biologic agent for the prevention of RCDI.
Methods: SER-109 is an ecology of bacterial spores enriched from stool from healthy, screened donors. The efficacy and safety of SER-109 was evaluated for prevention of RCDI in an open-label study of 30 outpatients with ≥ 3 episodes of CDI in the past 12 months who had responded to standard antibiotics. During 8 weeks following SER-109, 26/30 patients (87%) had no evidence of *C. difficile* related diarrhea, and 3 patients with transient diarrhea had resolution of symptoms without treatment intervention, for an overall clinical resolution of 96.7%. To determine the prevalence of VRE, baseline and week 4 stools were plated to Enterococcosel medium with 8 ug/mL vancomycin and titers of VRE (cfu/g) were determined.
Results: Of the 30 patients (67% female, mean age 66.5 years), 25 (83.3%) had been treated with vancomycin for RCDI. At baseline, 8 of 29 patients (27.6%) had VRE (ie, *E. faecium*) and in all 8 VRE titers decreased by 2-6 logs (to below the limit of detection) after SER-109. Other evidence of a remodeling of the microbiome by SER-109 includes a shift in the proportions of Enterobacteriaceae to a predominance of *E. coli,* and an increase in microbiome diversity detected via genomics analysis (Chao-1 diversity index of 24±8 at baseline shifted to 31±5 at 8 weeks, p < 0.01). Engraftment of species from SER-109 and augmentation (outgrowth) of non-spore forming bacteria (*Bacteroides* and *Parabacteroides*) not found in SER-109 were also observed.
Conclusion: VRE colonization was commonly observed among outpatients with RCDI. SER-109, an ecology of bacterial spores, was associated with significantly diminished VRE colonization. Restoration of the gut microbiome may be a novel infection control measure to provide colonization resistance against drug-resistant pathogens.

Background

- According to the Centers for Disease Control, an estimated 66,000 enterococcal infections occur among hospitalized patients in the US every year and about one-third are vancomycin-resistant [1]. Mortality secondary to bloodstream infections is more than doubled in patients with vancomycin-resistant *Enterococcus* (VRE) compared to patients with sensitive strains (2) and currently available therapeutic options for VRE have high treatment failure rates [2,3,4].
- Acquisition of VRE is associated with prior history of hospitalization and receipt of antibiotics, which are also risk factors for *Clostridium difficile* infection (CDI) [5,6]. Antibiotics disrupt the integrity of the microbiome leading to a low diversity state, which impairs the ability of the microbiome to defend against colonization by potential pathobionts, such as *C. difficile* [6,7]. Although vancomycin is commonly used to treat patients with recurrent CDI, relapse rates are high (>60%) and vancomycin exposure is associated with VRE colonization and expansion of the *Enterobacteriaceae* population [7, 8].
- In a recent phase 1b/2 trial, we assessed the efficacy and safety of SER-109, a novel microbiome agent, to prevent recurrent CDI in patients with a history of multiple relapses of CDI. SER-109 was developed as a microbiome-based therapy composed of spore-forming Firmicutes derived from the stool of healthy donors. In an animal model of recurrent CDI, spores were identified as the active component in preventing relapsing disease. Spore purification with ethanol selectively kills vegetative bacteria, fungi, parasites and most viruses, reducing the risk of pathogen transmission.
- In this early trial, we observed high rates of clinical efficacy, which occurred in parallel with increasing microbiome diversity towards levels observed in states of health [9]. We initiated a study to examine colonization by VRE and *Enterobacteriaceae* and determine whether SER-109 administration was associated with reductions in gut colonization in a high-risk patient population with a past history of heavy antibiotic exposure.

Methods

- In an open-label phase 1b/2 clinical study (SERES-001), 30 patients with a history of RCDI were enrolled in two different dosing cohorts. Patients were given SER-109 oral capsules 48 hours after completing therapy for CDI. The primary clinical endpoint was prevention of RCDI (>3 loose *C. difficile*-positive stools in 24 hours). The diversity of the GI microbiome was assessed pre-treatment with SER-109 and at weeks 1, 2, 4, 8 and 24.
- Titers (CFU/g feces) of vancomycin-resistant Enterococcus were determined by plating on Enterococcosel agar plus 8 ug/ml vancomycin. To positively identify isolates as VRE, colonies were purified, and MICs and species identity were determined via Vitek 2 cards (AST-GP70, and GP, respectively).
- Titers of *Bacteroides fragilis* group species and *Enterobacteriaceae* species were determined by plating to selective media (Bacteroides Bile Esculin agar, MacConkey lactose agar, respectively) and performing the following algorithm: titers of each colony morphology on each plate type were determined, and identification of multiple representatives of each colony type were determined by 16S Sanger sequencing. Species assignments were made using a phylogenetic approach, and titers for the species of interest were then calculated.

Results

Patient Demographics

Patients enrolled in the SERES-001 study must have experienced clinical improvement on their most recent antibiotic course of therapy. For twenty-three of the 30 patients (76.7%) the last antibiotic received was vancomycin.

Sex and Age

	Number	Mean Age	Stdev
Female	20	56.6	18.0
Male	10	68.0	15.0

Last Antibiotic Received and # Recurrences

	Number	Mean Recurrences	Stdev
fidaxomicin	5	3.2	1.1
metronidazole	1	2	N/A
rifaximin	1	4	N/A
vancomycin	23	3.3	1.1

Overview of SERES-001 Recurrent CDI Trial Results

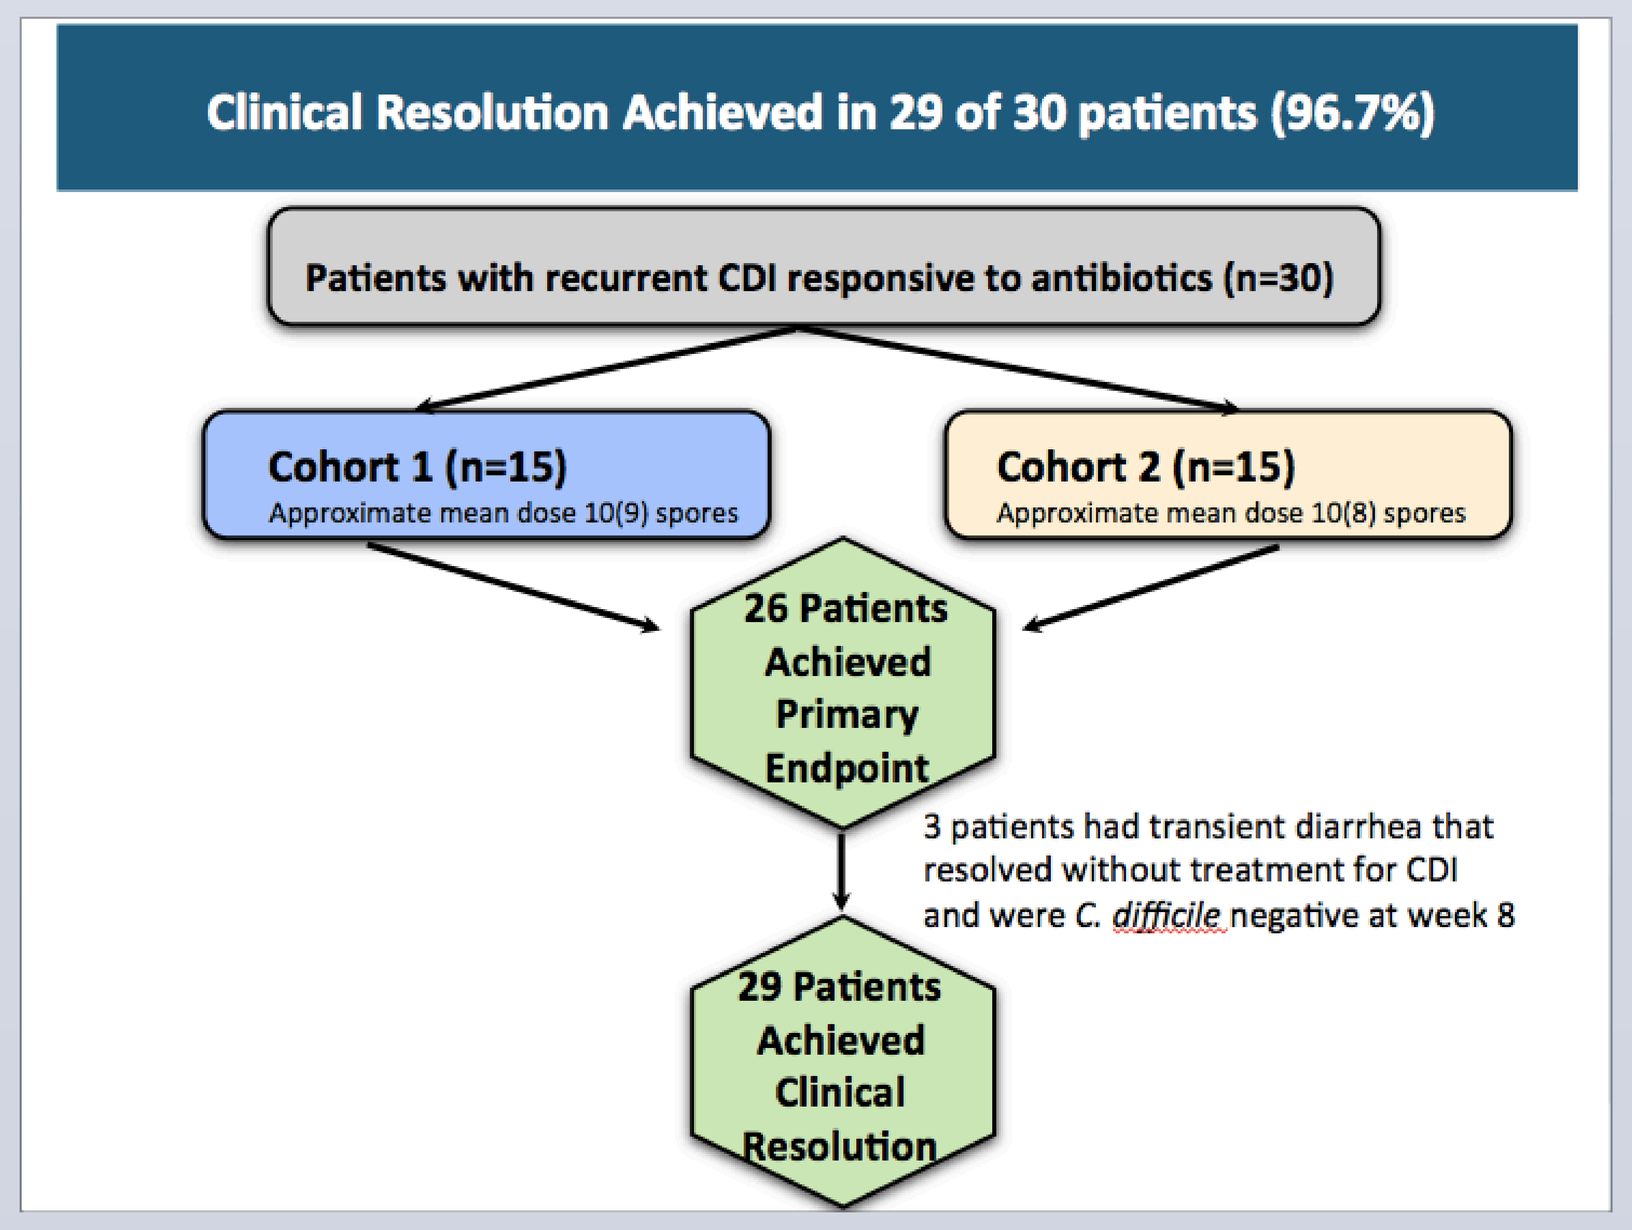

Clinical Resolution Achieved in 29 of 30 patients (96.7%)
Patients with recurrent CDI responsive to antibiotics (n=30)
Cohort 1 (n=15)
Approximate mean dose 10(9) spores
Cohort 2 (n=15)
Approximate mean dose 10(8) spores
26 Patients Achieved Primary Endpoint
3 patients had transient diarrhea that resolved without treatment for CDI and were C. difficile negative at week 8
29 Patients Achieved Clinical Resolution

Reduction of Vancomycin-resistant Enterococcus

Before receiving SER-109, 8 of 30 patients had evidence of VRE colonization with *E. faecium*; two patients were also colonized with *E. faecalis*. Expanded microbiome diversity post-treatment with SER-109 was associated with decreased VRE colonization and titers declined below the level of detection (LOD).

Pt	Pre-SER109	Wk 4	Fold reduction	Species
06R	2.7×10^5	$< 2.4\times10^2$	$> 1.1\times10^3$	*E. faecium*
07R	1.5×10^9	$< 1.6\times10^5$	$> 9.4\times10^3$	*E. faecium, E. faecalis*
08R	7.8×10^6	$< 1.0\times10^1$	$> 7.8\times10^5$	*E. faecium*
10R	1.1×10^7	$< 1.0\times10^5$	$> 1.1\times10^2$	*E. faecium, E. faecalis*
12R	2.5×10^8	$< 1.6\times10^2$	$> 1.4\times10^6$	*E. faecium*
13R	1.2×10^9	$< 1.0\times10^5$	$> 1.2\times10^4$	*E. faecium*
04RDD	4.6×10^8	$< 5.0\times10^5$	$> 9.2\times10^2$	*E. faecium*
14RDD	8.7×10^7	$<1.0\times10^4$	$> 8.7\times10^3$	*E. faecium*

LOD varies due to background of other species on selective agar. MICs were confirmed to be >32 ug/ml for at least 1 isolate from each patient.

Enterobacteriaceae are remodeled post-treatment with SER-109

Changes in the *Enterobacteriaceae* were detected post-treatment with SER-109 via culture-based methods. The number of *Enterobacteriaceae* species was significantly reduced (left) and the fraction of *E. coli* out of the total *Enterobacteriaceae* was significantly increased (right).

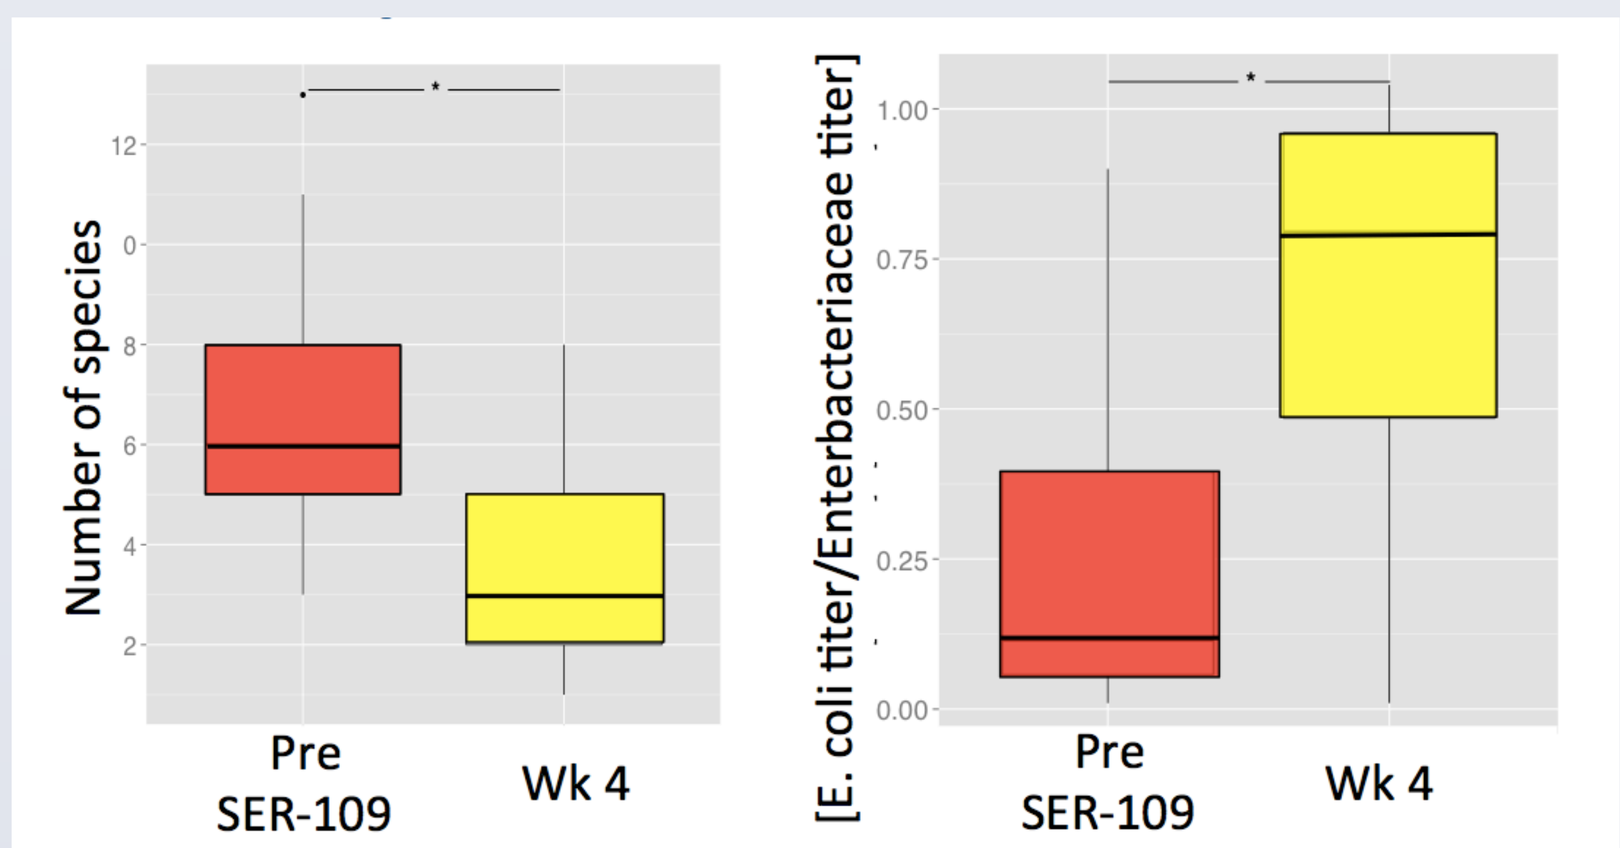

Number of species
Pre SER-109
Wk 4
[E. coli titer/Enterbacteriaceae titer]
Pre SER-109
Wk 4

Difference between species number pre- and post-treatment, p < 0.0001. Change in *E. coli* as a fraction of the total *Enterobacteriaceae*, p = 0.0002. p values, Wilcoxon signed-rank test.

Treatment with SER-109 was associated with increased bacterial diversity in the gastrointestinal microbiota

The increased diversity of bacteria post-treatment with SER-109 is presented below. The Chao-1 alpha diversity index (top) presents an estimate of the number of distinct bacterial clades in stool samples from patients pre-treatment and at week 8 post-treatment, and in stool samples from donors (the 7 healthy volunteers who provided stool for SER-109 processing). The principal components plot (bottom) indicates the distinctness of pre-treatment samples and a post-treatment trend towards similarity to stool samples from healthy volunteers.

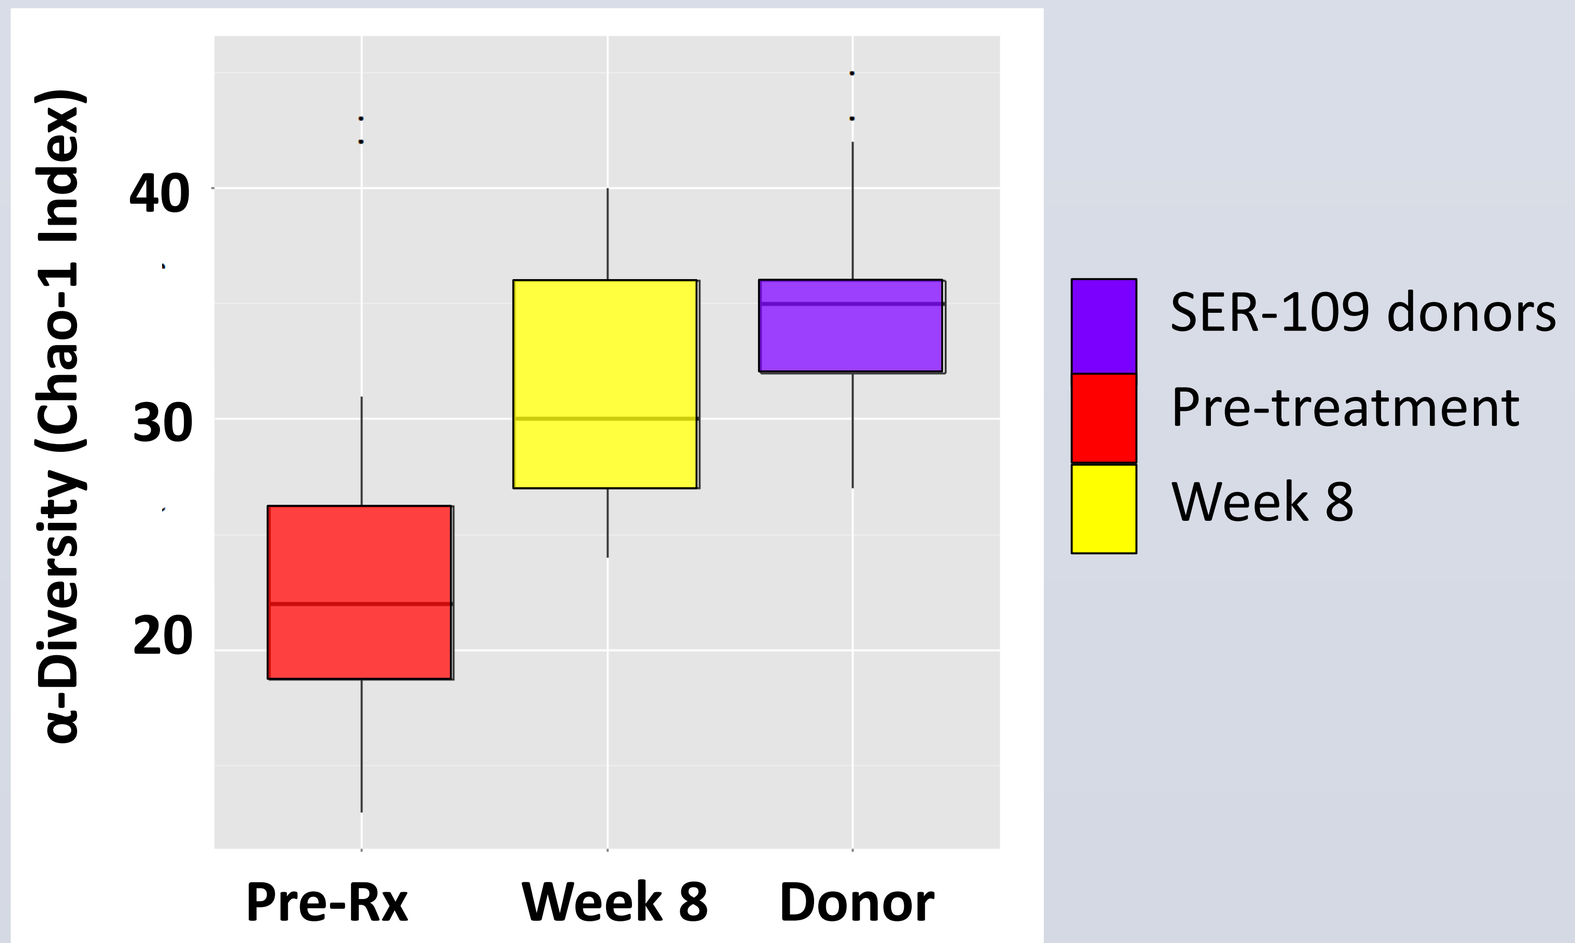

α-Diversity (Chao-1 Index)
40
30
20
Pre-Rx
Week 8
Donor
SER-109 donors
Pre-treatment
Week 8

Average microbiome alpha-diversity in patients pre- and post-treatment with SER-109. Diversity indices are calculated based on taxonomic assignments to phylogenetic clades. All pairwise comparisons are significant at p<0.001 (Wilcoxon signed-rank test).

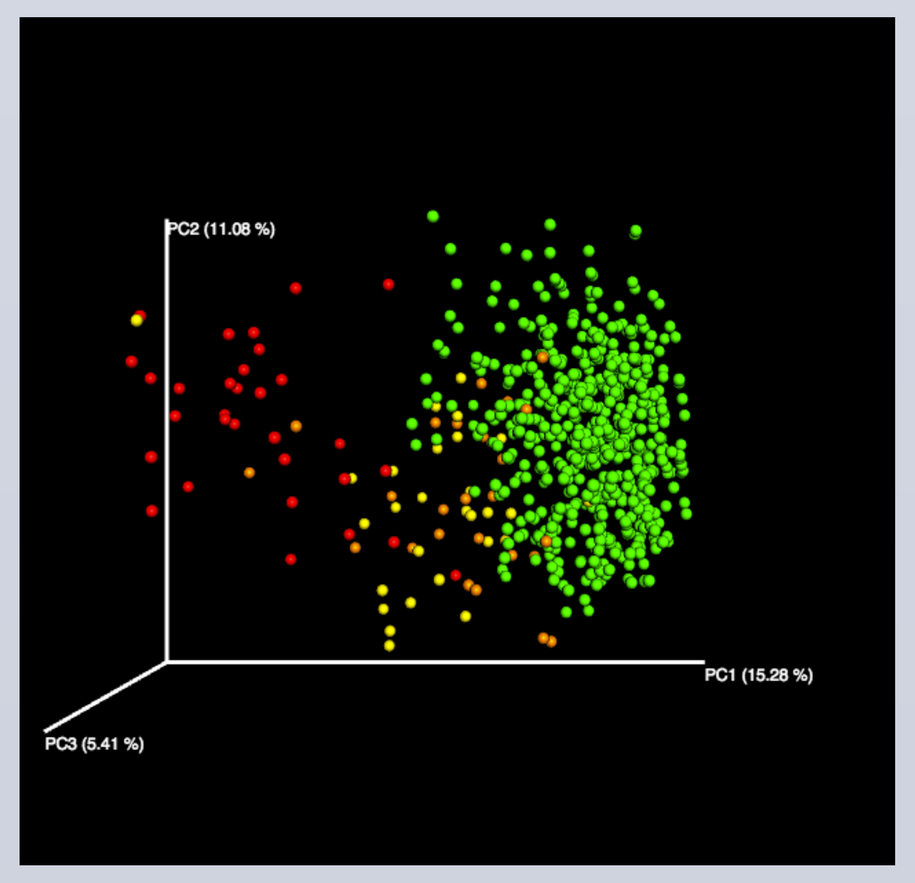

PC2 (11.08 %)
PC1 (15.28 %)
PC3 (5.41 %)

HMP = green
Pre-treatment = red
8-16 wks = yellow
24-27 wks = orange

Principle Coordinates Analysis shows comparability of GI microbiome of patients at ≥ 8 weeks post-treatment with SER-109 to microbiome of healthy individuals sampled as part of the Human Microbiome Project based on unweighted UniFrac analysis.

Increased diversity was achieved via *engraftment* of SER-109 species and *augmentation* of species not present within SER-109.

Engraftment was defined as species that were detected in SER-109, and detected post-treatment in patients, but not detected pre-treatment. The top figure below presents engraftment data for all patients, based on genomics analyses and coded to identify SER-109 processed from each of the 7 donors in the study. In all patients, multiple phylogenetic clades of organisms are found to engraft and engraftment is independent of the dose of spores. The bottm figure presents augmentation data from culture-based analysis of the *B. fragilis* group. Augmentation was defined as species not present in SER-109 that are detected at greater than 10-fold higher relative abundance post-treatment than pre-treatment. In 11 of 29 patients, the CFU/g of *Bacteroides* and *Parabacteroides* in stool were increased by 38 to 1.2 million fold.

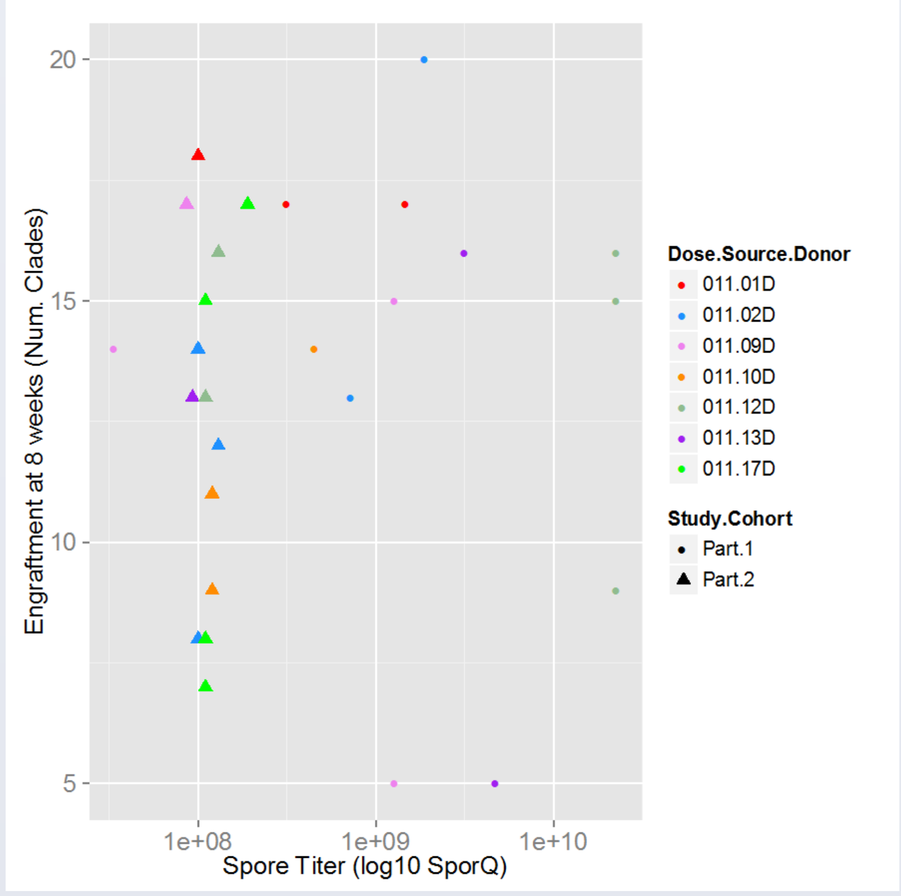

Engraftment at 8 weeks (Num. Clades)
20
15
10
5
1e+08
1e+09
1e+10
Spore Titer (log10 SporQ)
Dose.Source.Donor
011.01D
011.02D
011.09D
011.10D
011.12D
011.13D
011.17D
Study.Cohort
Part.1
Part.2

Top: Engraftment of phylogenetic clades in SER-109. The number of engrafted clades for each patient is shown, coded by donor and by cohort. The dose of SER-109 is indicated on the X-axis.

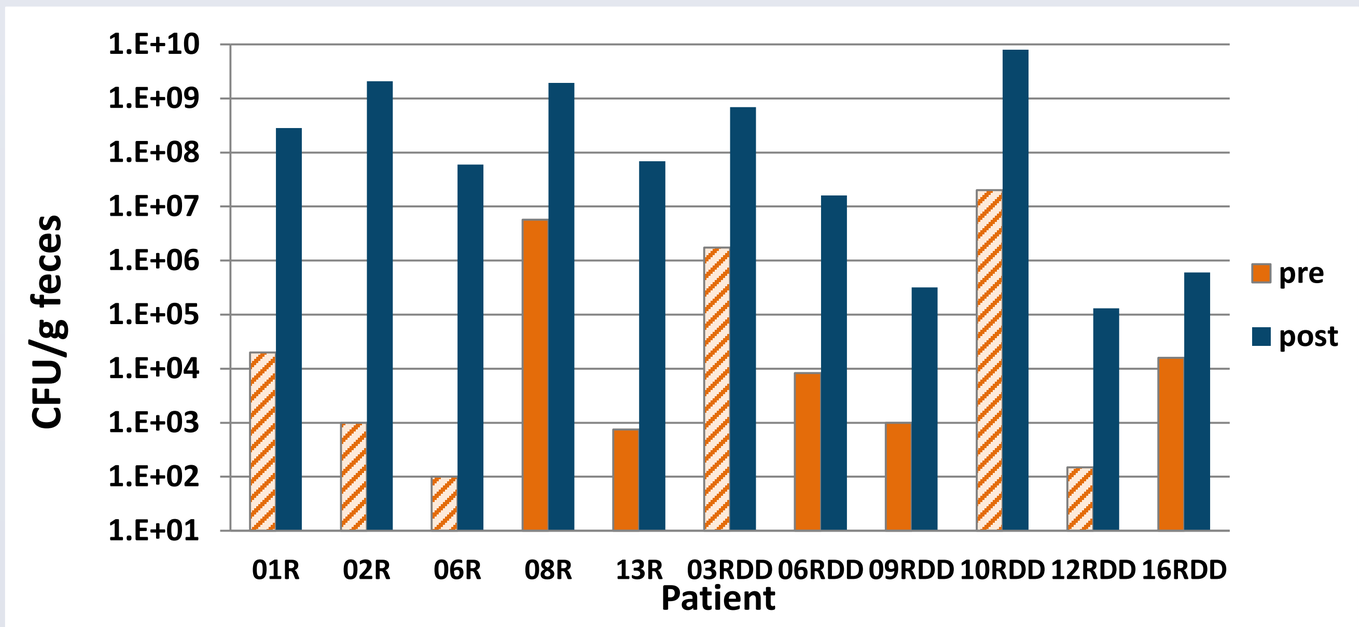

CFU/g feces
1.E+10
1.E+09
1.E+08
1.E+07
1.E+06
1.E+05
1.E+04
1.E+03
1.E+02
1.E+01
01R
02R
06R
08R
13R
03RDD
06RDD
09RDD
10RDD
12RDD
16RDD
Patient
pre
post

Bottom: Augmentation of Bacteroides and Parabacteroides. Carriage pre- and post-treatment with SER-109 shown in CFU/g feces. Hatched bars, not detected

Summary

- In the 8 patients in whom VRE was detected at baseline, VRE titers were reduced to below the limit of detection post-treatment with SER-109.
- *Enterobacteriaceae* species number was reduced post-treatment and the population shifted towards an *E. coli* dominated community, typical of healthy individuals.
- The diversity of the gut microbiome of patients increased significantly post-treatment with SER-109 and the overall microbiome ecology became more similar to healthy individuals.
- SER-109 species engrafted and non-SER-109 species found in healthy GI microbiomes, such as *Bacteroides* and *Parabacteroides*, were augmented.

Conclusions

SER-109 treatment was associated with a high rate of clinical resolution of recurrent CDI. In parallel, SER-109 led to increased diversity of the microbiome in patients with a history of recurrent CDI, who generally have a low diversity state secondary to repeated antibiotic exposure.
Remodeling of the microbiome was associated with other benefits including decreased colonization with VRE and lower proportions of potential pathobionts within the *Enterobacteriaceae* family. These data suggest that remodeling of the gut microbiome with SER-109 may decrease carriage of VRE and other drug-resistant organisms, a hypothesis that will be explored in our ongoing Phase 2 trial.

REFERENCES
1. DHHS/CDC, Antibiotic Resistance Threats in the US, 2013
2. DiazGranados CA, JID 2005
3. Mermel LA, Clin Infect Dis 2009
4. Britt, Clin Infect Dis 2015
5. Furuno, Arch Int Med 2006
6. Loo VG, N Eng J Med 2011
7. Lewis BB, J Infect Dis 2015
8. Van Nood, NEJM 2013
9. Pardi DS, B-1875a presentation, ICAAC 2014

RESEARCH POSTER PRESENTATION DESIGN © 2015
www.PosterPresentations.com